2018 ASA Biopharmaceutical
Section Regulatory-Industry
Statistics Workshop
Poster No. 300685

Considerations on Group Sequential Design for Multiple Endpoints with Different Information Fractions

Dongyuan Xing, Jane Wu, Matthew Guo; *Eisai Inc.*

Introduction

Clinical trials often include a family of endpoints as indicators of treatment effects. The family wise error rate (FWER) is the probability of erroneously finding a statistically-significant treatment effect in at least one endpoint regardless of the presence or absence of treatment effects in the other endpoints within the family. A statistical strategy in a fixed sample size (or event size) setting, testing endpoints in a predefined hierarchical order, all at the same significance level alpha (e.g., α = 0.05 two-sided) and moving to the next endpoint only after a success on the previous endpoint, does not inflate FWER. However, this hierarchical testing strategy may not maintain a control of FWER in the setting when there are interim analyses, e.g., group sequential design (GSD). This problem was first studied by Hung, Wang and O'Neill (2007).[1]

We consider a clinical trial of a randomized two-arm (1:1 randomization ratio) study where a test arm is compared to a control arm for superiority on a primary endpoint (E1) and a secondary endpoint (E2), where the secondary endpoint is tested only if testing of the primary endpoint is a success. Hung, Wang and O'Neill (2007)[1] investigated three strategies for testing the secondary endpoint in this setting.

- Strategy 1 — test the secondary endpoint with full alpha (e.g., one-sided 0.025) after the null hypothesis for the primary endpoint is rejected;
- Strategy 2— test the secondary endpoint using the same rejection boundary of the primary endpoint;
- Strategy 3 —test the secondary endpoint with critical value of $Z_{\alpha/2}$.

Hung, Wang, O'Neill (2007) concluded that Strategy 1 may inflate type I error rate of secondary endpoint and the maximum type I error occurs when the effect size of primary endpoint is moderate and correlation of E1 and E2 is one; Strategy 2 can control Type I error rate of secondary endpoint, however, it may be too conservative; Strategy 3 controls type I error rate and can be substantially more powerful than Strategy 2.

In the same setting, Tamhane , Mehta and Liu (2010) found that, using any alpha-level boundary of a spending function for testing secondary endpoint conditional on rejecting the null hypothesis of primary endpoint, the overall FWER will be controlled. They studied four combinations consisting of O'Brien–Fleming (OF) and Pocock (PO) boundaries for the primary and secondary endpoints (i.e., OF1 –PO2, OF1-OF2, PO1-PO2, and PO1-OF2). The OF1-PO2 combination is recommended as it generally gives higher power for both the primary and secondary endpoints compared with the other strategies.[2,3]

Glimm, Maurer and Bretz (2010) independently obtained the same results, they also considered other variations of settings such as two primary endpoints instead of one primary and one secondary.[4]

Li, Wulfsohn and Koch (2017) considered settings with more than one interim analysis and more than one secondary endpoint. They proposed an 'adjusted Pocock approach" in which less stringent boundary for secondary endpoints is identified by trial and error method under OF1-PO2 structure.[5]

In all references above, the information fraction at interim analysis is assumed to be the same for primary and secondary endpoints. We investigated the phenomenon in the setting where information fraction is different for the primary and secondary endpoints. Our research is motivated by phase 3 oncology trials in which time-to-event endpoint overall survival (OS) is often chosen as primary endpoint and progression-free survival (PFS) as secondary endpoint or vice versa, the timing of interim analysis is determined based on the information fraction of the primary endpoint and the maturity of secondary endpoint at interim analysis could be different from that of the primary endpoint. We evaluate how the endpoint-specific information fractions impact the type I error rate and power for secondary endpoint under different treatment effects on the primary endpoint and different correlations between endpoints.

Methods

Consider scenario with one primary endpoint E1 and one secondary endpoint E2. One interim analysis and one final analysis are planned. The timing of interim analysis is determined by the information fraction of E1 while the information fraction of E2 could be different from that of E1. The information fraction at the final analysis is assumed to be 100% for both E1 and E2. E2 is only tested when the null hypothesis for E1 is rejected.

Denote the null hypotheses in testing E1 and E2 as H^0_i: $S_i(t)=S^0_i(t)$ (i=1, 2), where $S_i(t)$ and $S^0_i(t)$ are survival functions of the i^{th} endpoint in test arm and control arm, respectively; and Type I error rate is one-sided level of 0.025; denote HR_i i=1, 2 as hazard ratio (HR) for the i^{th} endpoint of test arm over control arm; denote the standardized test statistics as Z_{ij} for i^{th} endpoint at interim analysis (j=1) and final analysis (j=2); denote the information fraction as t_{ij}, where $0<t_{i1}<1$ and $t_{i2}=1.0$; and denote critical boundaries as (c_{i1}, c_{i2}) for i^{th} endpoint at j^{th} time point.

The FWER under configuration of intersection of null hypotheses $H^0_1 \cap H^0_2$ is controlled at α level [2]. Here, we evaluate FWER under different H_1 (i.e. different treatment effect on E1) and null hypothesis of H^0_2. Therefore, the FWER is equivalent to the type I error rate of testing E2. The type I error rate (one-sided) of testing E2 is defined as

$$\alpha_2 = \Pr(Z_{11} > c_{11}, Z_{21} > c_{21} | H^0_2) + \Pr(Z_{11} < c_{11}, Z_{12} > c_{12}, Z_{22} > c_{22} | H^0_2) \qquad (1)$$

For a time-to-event variable, the joint distribution of group sequentially computed log-rank test statistics converges asymptotically to multivariate normal distribution when proportional hazard assumption holds [6]. So, the standardized test statistics at the interim and final analyses (Z_{i1}, Z_{i2}) for the i^{th} endpoint E_i follow bivariate normal distribution with mean $(\log(HR_i)\sqrt{I_{i1}}, \log(HR_i)\sqrt{I_{i2}})$ and correlation as $\mathrm{corr}(Z_{i1}, Z_{i2}) = \sqrt{t_{i1}}$ and $\sqrt{I_{i1}} \approx \sqrt{\frac{D_i}{4} t_{i1}}$ where D_i =the number of observed events at final analysis.

The joint distribution of $(Z_{11}, Z_{12}, Z_{21}, Z_{22})$ is assumed four-variate normal with correlation given as below [5]

$$corr(Z_{i1}, Z_{i2}) = \sqrt{t_{i1}}, \quad corr(Z_{12}, Z_{22}) = \rho, \quad corr(Z_{11}, Z_{22}) = \rho\sqrt{t_{i1}}$$
$$corr(Z_{11}, Z_{21}) = \rho\sqrt{\frac{t_{11}}{t_{12}}}\ (\text{if } t_{11} < t_{21}) \text{ and } corr(Z_{11}, Z_{21}) = \rho\sqrt{\frac{t_{21}}{t_{11}}}\ (\text{if } t_{21} \le t_{11}) \qquad (2)$$

Note that ρ is the correlation of the E1 and E2 at final analysis and the variance of all four test statistics Z_{ij} (i=1, 2; j=1, 2) is equal to 1.

In the settings designed above, we evaluate the performance of different E2 boundary selection approaches in terms of type I error and power; and propose how to select E2 boundaries in the situation that primary and secondary endpoints having different information fractions. Type I error and power were calculated using numerical integration methods in R. Simulations were run to evaluate the assumptions on correlation matrix. The Lan-DeMets alpha-spending function with OF or Pocock type approximation was used.

Results

Figure 1. Type I error rate of E2 under different information fractions of E1 and E2 when (c_{11}, c_{12}) = OF boundary and E2 is tested at one-sided 0.025 level (full alpha) whenever E1 achieves significance—Strategy A.

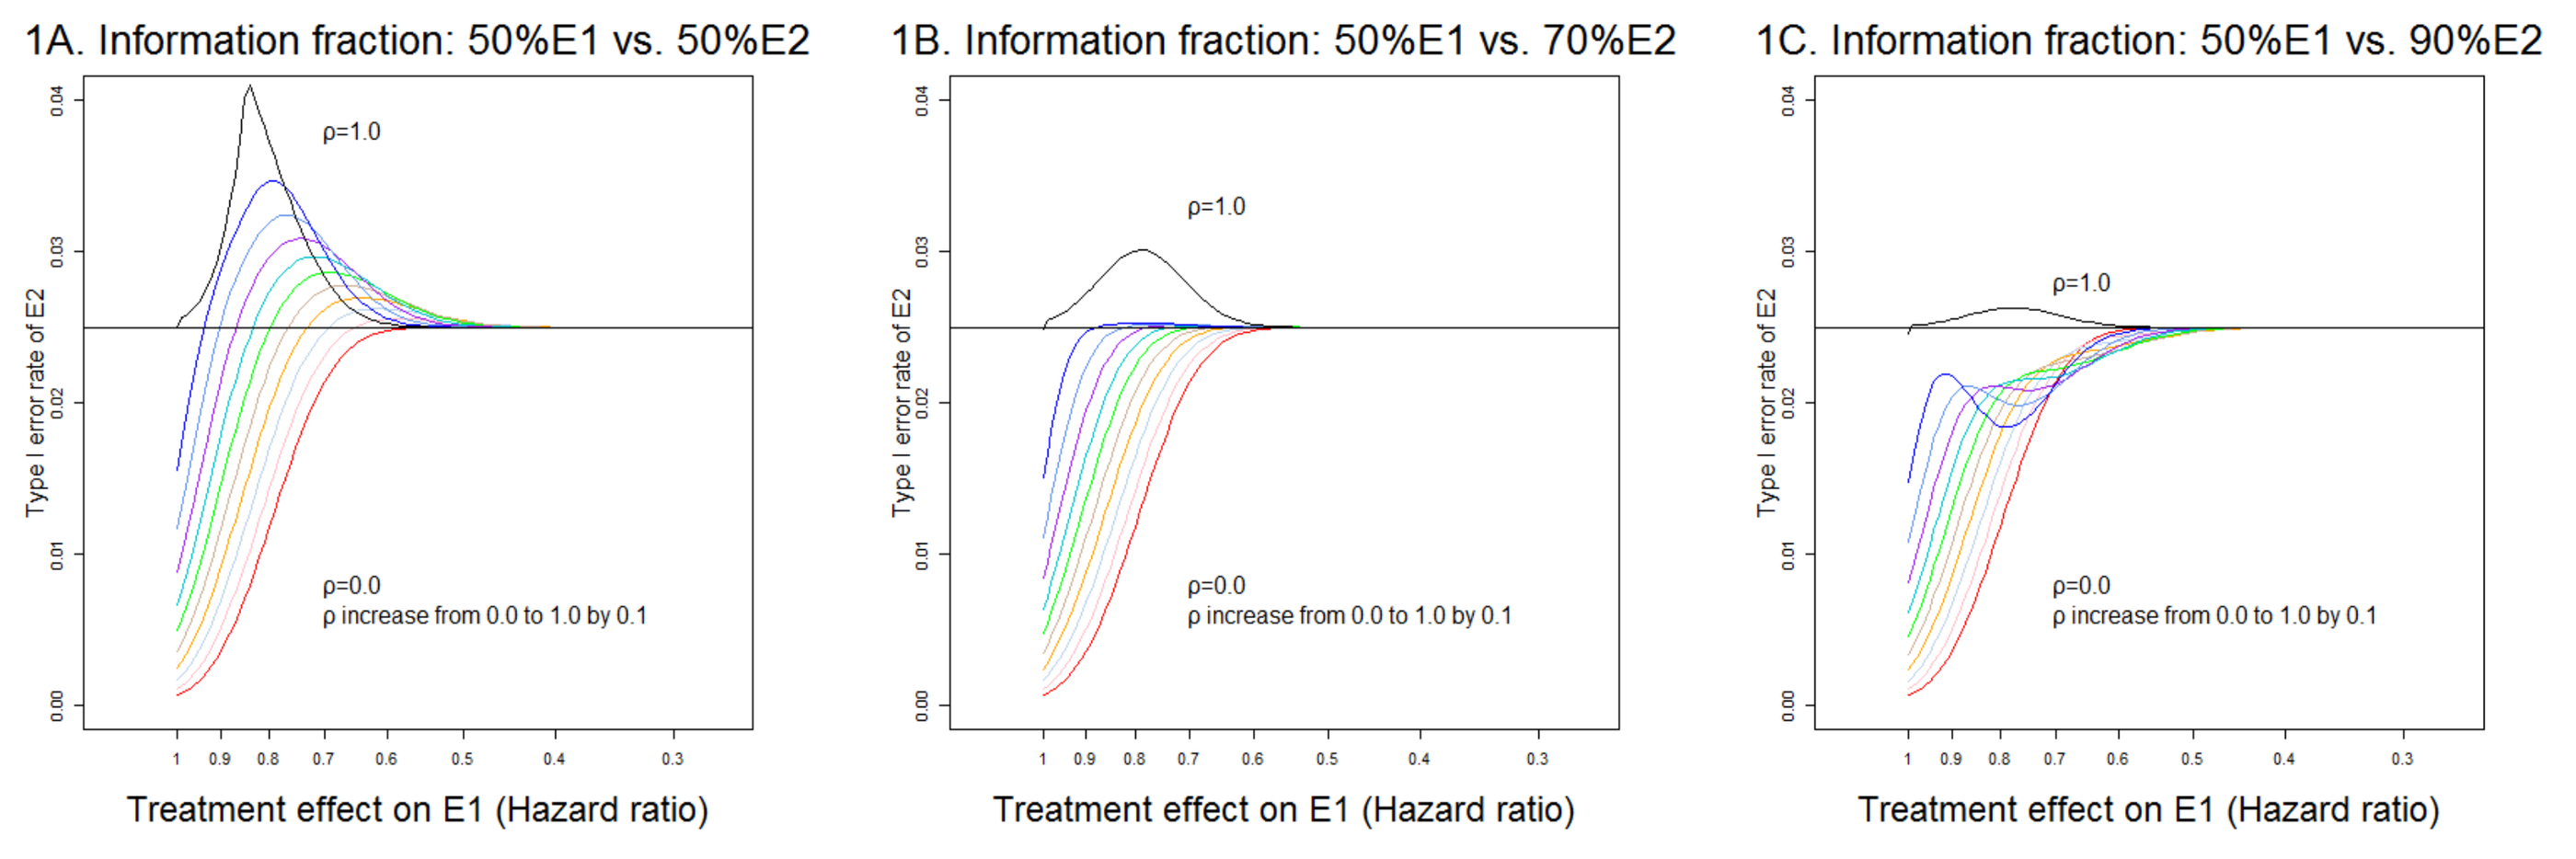
1A. Information fraction: 50%E1 vs. 50%E2
1B. Information fraction: 50%E1 vs. 70%E2
1C. Information fraction: 50%E1 vs. 90%E2
ρ=1.0
ρ=0.0
ρ increase from 0.0 to 1.0 by 0.1
Type I error rate of E2
Treatment effect on E1 (Hazard ratio)

- OF boundaries at one-sided 0.025 level for E1: c_{11}=2.963, c_{12}=1.969; boundaries for E2: $c_{21}=c_{22}$=1.960

Results (cont'd)

- Figure 1A, 1B and 1C confirm the finding by Hung, Wang and O'Neill (2007) [1] on Strategy 1 under varying information fractions of E1 and E2. However, the inflation in α_2 becomes less concerned when the difference in the information fractions of E1 and of E2 gets larger. Under the scenario of 50% information fraction for E1 and 90% for E2, inflation is observed when ρ=1.0.

Figure 2. Type I error rate of E2 under different information fractions of E1 and E2 when (c_{11}, c_{12}) = OF boundary and (c_{21}, c_{22}) = PO boundary and E2 is tested whenever E1 achieves significance —Strategy B.

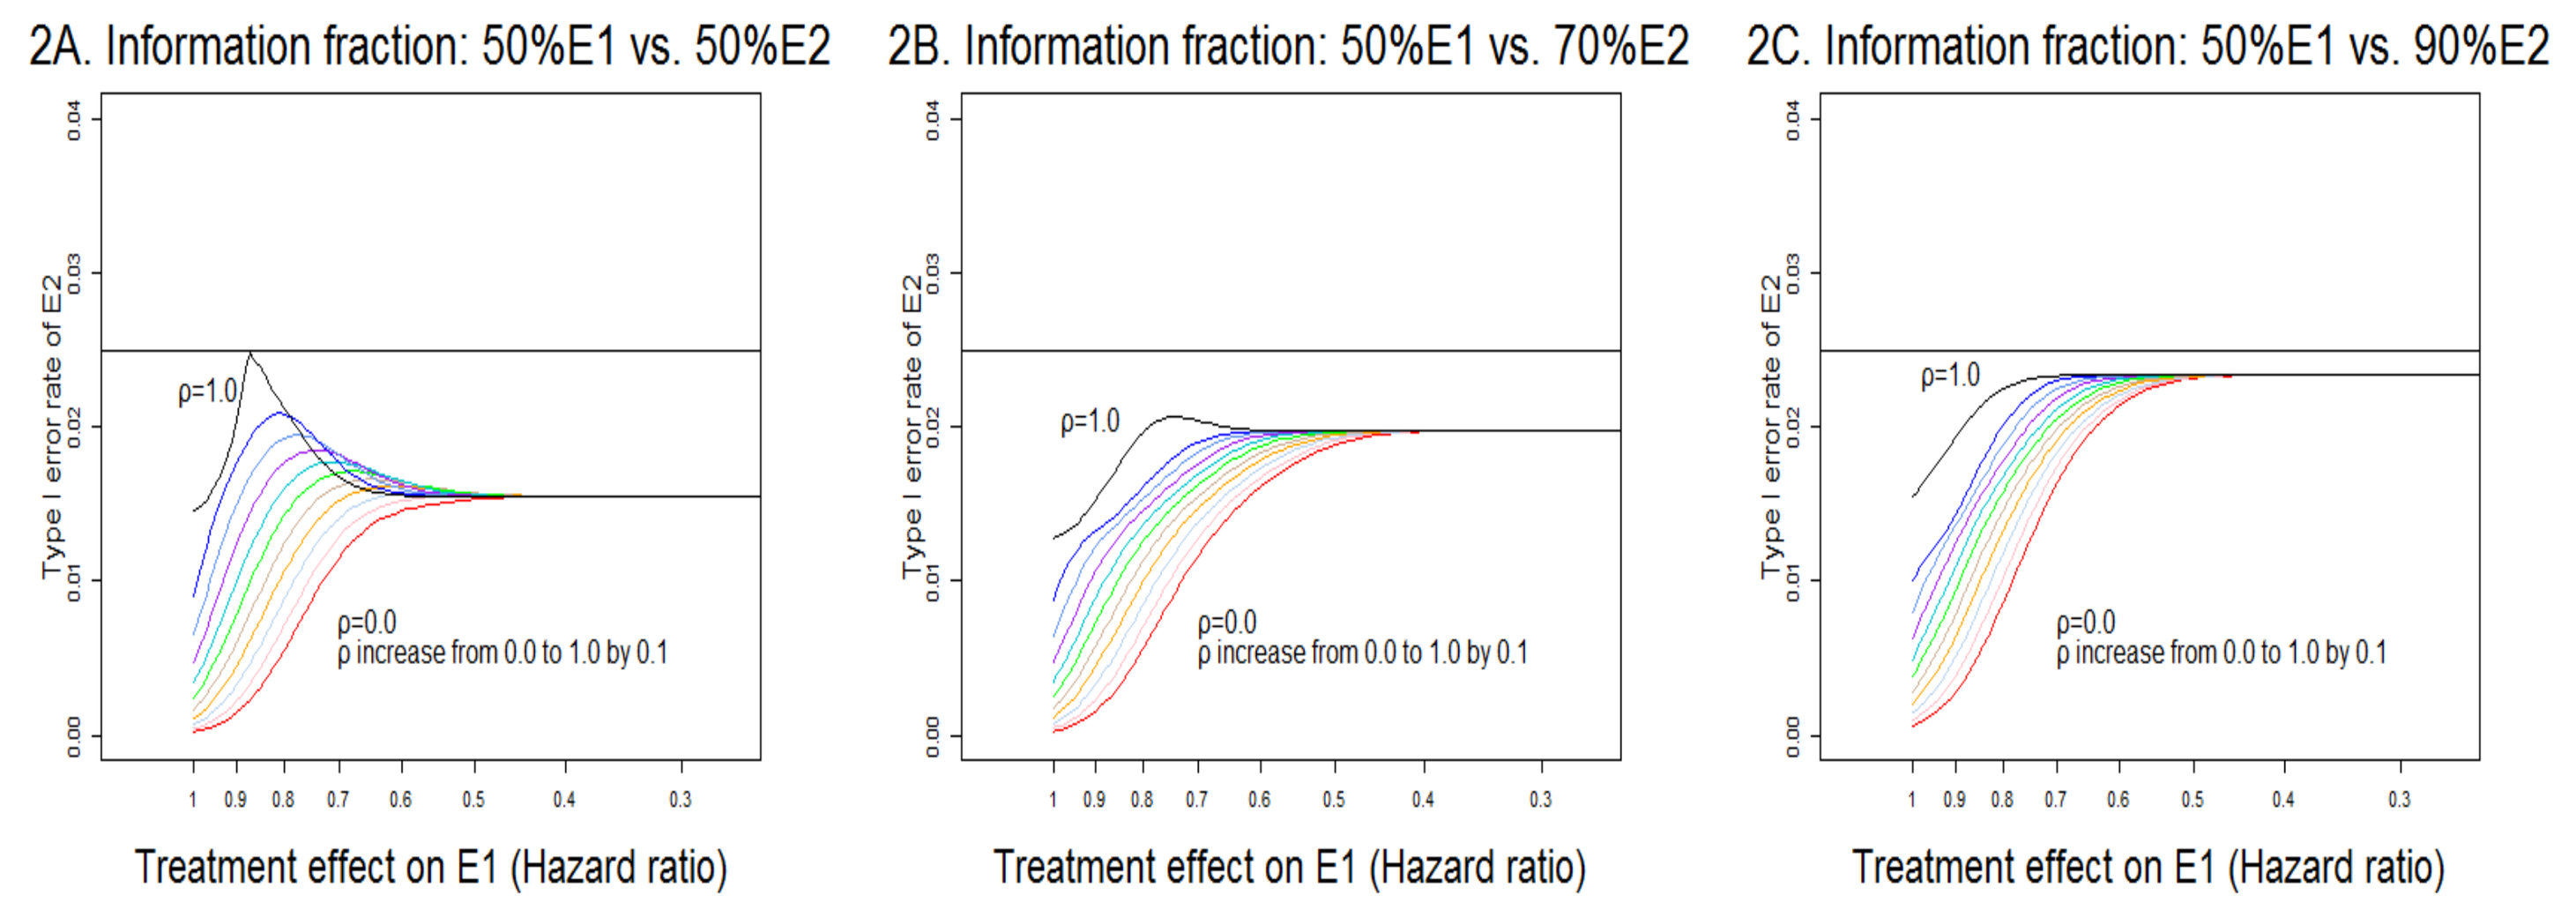
2A. Information fraction: 50%E1 vs. 50%E2
2B. Information fraction: 50%E1 vs. 70%E2
2C. Information fraction: 50%E1 vs. 90%E2
ρ=1.0
ρ=0.0
ρ increase from 0.0 to 1.0 by 0.1
Type I error rate of E2
Treatment effect on E1 (Hazard ratio)

- OF1 boundaries: (2.963, 1.969); PO boundaries: (2.16, 2.20) in 2A, (2.06, 2.25) in 2B, (1.99, 2.25) in 2C.
- The strategy of OF1-PO2 is recommended by Tamhane, Mehta and Liu (2010)[2] . As depicted in Figure 2, the approach controls maximum α_2 =0.025 over a rang of ρ when information fraction is the same for E1 and E2 (Figure 2A), but maybe too conservative as maximum α_2 for correlation ρ<0.5 is well below 0.025.
- When the difference in information fractions increases as shown from 2A to 2B and from 2B to 2C, the maximum α_2 gets closer to 0.025 for all levels of correlation ρ, which means over-conservativeness of Strategy OF1-PO2 is of a less concern; and when the difference in information fractions is big, it is appropriate to use the strategy OF1-OP2.

Table 1. Keep OF boundary for E1 and find adaptive boundaries for E2 $c_{21}=c_{22}$ that maximize α_2 =0.025 (OF1-AD2) — Strategy C.

OF1-AD2	50%E1 vs. 50%E2	50%E1 vs. 70%E2	50%E1 vs. 90%E2
ρ=0	1.960	1.956	1.955
ρ=0.6	2.033	1.955	1.956
ρ=1.0	2.173	2.040	1.981

- Table 1 includes boundary values for E2 ($c_{21}=c_{22}$) that maximize type I error rate α_2 at a given one-sided level of 0.025 in a practically meaningful range of hazard ratio of (0.6-1.0) for E1. The values can be found by solving α_2 =0.025 in (1).

Figure 3. Power of E2, comparison of Strategy B and C under different scenarios of information fraction of E1 and E2.

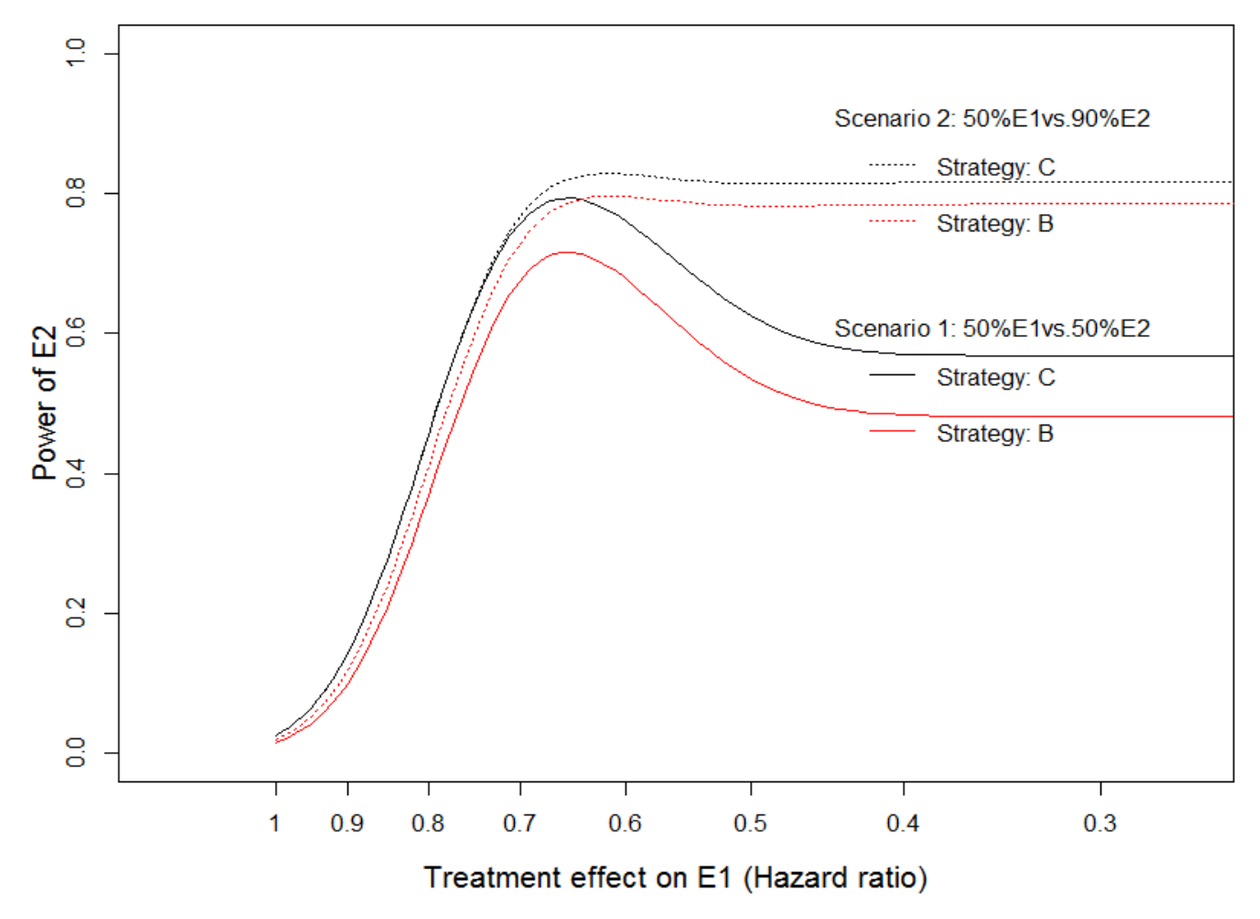
Scenario 2: 50%E1vs.90%E2
Strategy: C
Strategy: B
Scenario 1: 50%E1vs.50%E2
Strategy: C
Strategy: B
Power of E2
Treatment effect on E1 (Hazard ratio)

- In Figure 3, ρ=0.6, HR_2=0.7 and $D_1=D_2$=300.
- Strategy C is more powerful than Strategy B under both scenarios: 1) 50% E1 vs. 50% E2; and 2) 50% E1 vs.90% E2. Strategy A is not included as it does control type I Error rate in all scenarios.
- When treatment effect on E1 is large (e.g., HR_1<0.6), power of E2 is greater in Scenario 2 than in Scenario 1. This is reasonable because when treatment effect on E1 is large, it is more likely to test E2 at interim analysis, so power of E2 is larger if more data (90% information fraction) was observed at interim analysis.

Discussions

- We evaluate the performance of different E2 boundary selection strategies in the setting that the timing of interim analysis is determined by the information fraction of E1 while the information fraction of E2 could be different from that of E1. The O'Brien–Fleming (OF) boundary for the primary endpoint E1 was used in all strategies.
- We found that when the information fractions of E1 and of E2 at interim analysis are close, Strategy A, testing E2 at 0.025 level when E1 is rejected, may cause inflation in type I error rate of E2 (α_2). Strategy B (OF1-PO2) with Pocock boundaries for E2 may be considered. However, Strategy B may be too conservative when correlation is less than 1 and the treatment effect on E1 is small (Figure 2A). Strategy C, which use numerical approach to find (c_{21}, c_{22}) that maximize type I error under a given alpha level, is more powerful than Strategy B (Figure 3).
- When information fractions of E1 and E2 are very different (Figure 1C), the inflation in α_2 with Strategy A becomes minimal. Strategy A may be reasonable to use as long as correlation is not close to 1.
- Results presented here focus on scenarios where information fraction of E2 is larger, as our research was motivated by a phase 3 trial with OS as the primary and PFS as the secondary endpoint. The methods in this presentation can be easily extended to settings with information fraction of E1 is larger.
- The correlation matrix in (2) can be derived if two endpoints (E1 and E2) follow bivariate normal distribution. In our research, we made assumptions on the correlation of two test statistics associated with testing of E1 and E2 instead of correlation of two endpoints (E1 and E2) because it is not straightforward to assess correlation of time-to-event endpoints with censoring. To verify the validity of the assumptions, we run simulations to investigate the correlation of test statistics when PFS and OS follow exponential distribution using methods by Fleischer et al[8] . We found that when the hazard ratios of two endpoints are similar and ratio of median PFS is relatively larger, our assumptions appear very reasonable.
- In a clinical trial design, it will be useful to use simulation to estimate the correlation matrix of two endpoints with assumptions in the study design and use the correlation from the simulation to re-evaluate the E2 boundary selection strategies.

References

1. J Hung, S Wang and R O'Neill (2007). Statistical Considerations for Testing Multiple Endpoints in Group Sequential or Adaptive Clinical Trials. *Journal of Biopharmaceutical Statistics*, 17:6, 1201-1210
2. A Tamhane, C Mehta and L Liu (2010). Testing a Primary and a Secondary Endpoint in a Group Sequential Design. *Biometrics* 66, 1174–1184
3. A Tamhane, Y Wu and C Mehta (2012). Adaptive extensions of a two-stage group sequential procedure for testing primary and secondary endpoints (I): unknown correlation between the endpoints. *Statistics in Medicine* 31, 2027–2040
4. E Glimm, W Maurer and F Bretz (2010). Hierarchical testing of multiple endpoints in group sequential trials. *Statistics in Medicine* 29, 219–228.
5. X Li, M Wulfsohn and G Koch (2017). Considerations on Testing Secondary Endpoints in Group Sequential Design. *Statistics in Biopharmaceutical Research* 9:4, 333-337
6. AA Tsiatis (1981). The asymptotic joint distribution of the efficient scores test for the proportional hazards model calculated over time. *Biometrika*, **68**, 311–315
7. C Jennison and B Turnbull (2000). Group Sequential Methods with Applications to Clinical Trials. New York: Chapman and Hall/ CRC.
8. F Fleischer, B Gaschler-Markefski and E Bluhmki (2009). A statistical model for the dependence between progression-free survival and overall survival *Statistics in Medicine* **28**: 2669–2686.

Eisai Inc.
155 Tice Blvd
Woodcliff Lake, NJ 07677