Meta Analysis for All-cause Death for Apixaban vs. Placebo Or Control in SPAF Trials

Li Wang, Ph.D, Weihua Tang, Ph.D

Bristol-Myers Squibb Company, Hopewell, NJ 08534

BACKGROUND

•Apixaban was superior to warfarin for reduction of all-cause death in ARISTOTLE SPAF trial[1]

•Apixaban reduced the incidence of all-cause death comparing to aspirin (ASA) although it was not statistically significant in AVERROES SPAF trial[2]

•It is of great interest to identify the effect of apixaban vs. placebo or control (other than warfarin and ASA) in terms of reducing all-cause death in SPAF trials

DATA AVAILABLITY

•Apixaban vs. warfarin[1]
•Apixaban vs. ASA[2]
•Warfarin vs. placebo or control [3]
•ASA vs. placebo or control[3]

METHODOLOGY

•Indirect Comparison:
- •When head to head direct comparison data is not available, indirect comparison is a very useful tool to make inferences
- •Naïve indirect comparison should be avoided[4]
- •Adjusted indirect comparison usually gives consistent results with direct comparison [5]

•Meta Analysis:
- •When there are more than one trial with the same comparators, Meta-analysis is often used to combine findings from these independent trials to provide a more precise estimate of the treatment effect, weighted by the size of the studies
- •Heterogeneity between trials should be explored

PROPOSED ANALYSIS DIAGRAM

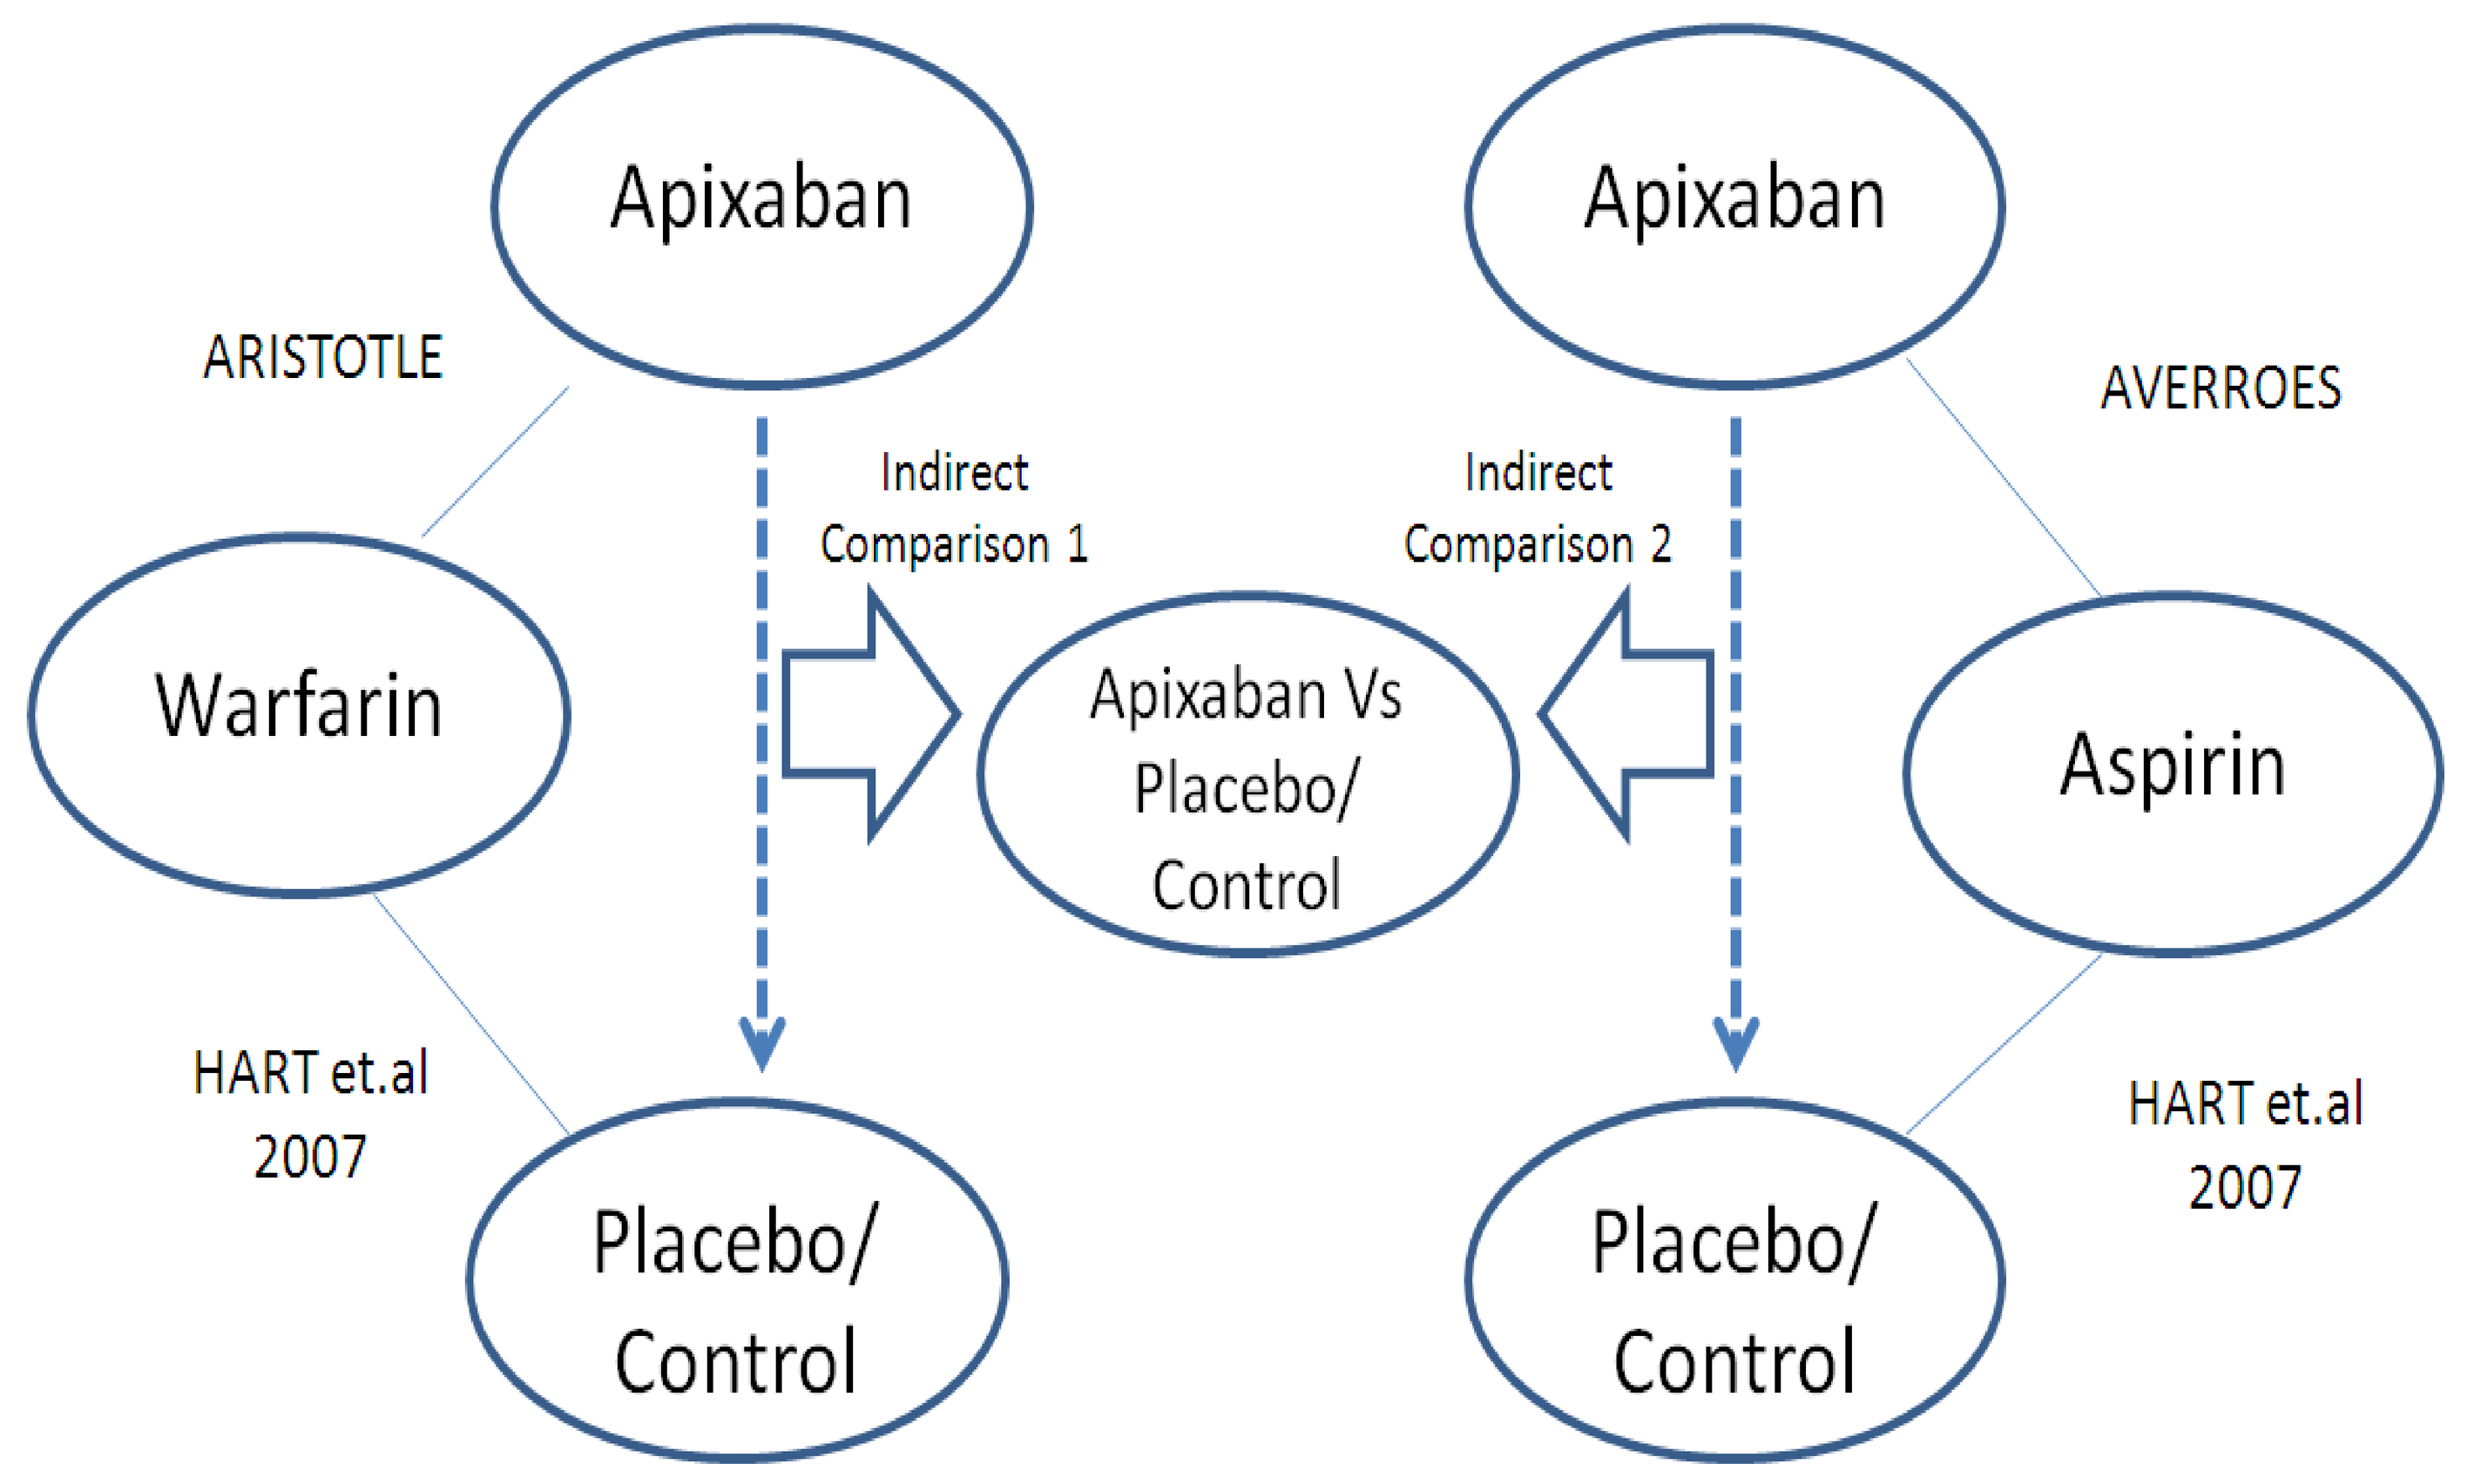
Apixaban
ARISTOTLE
Warfarin
HART et.al 2007
Placebo/ Control
Indirect Comparison 1
Apixaban Vs Placebo/ Control
Indirect Comparison 2
Apixaban
AVERROES
Aspirin
HART et.al 2007
Placebo/ Control

STATISTICAL ANALYSIS DETAILS

•Indirect Comparison for Odds Ratio:

$$\ln(OR_{\mathrm{Ind}}) = \ln(OR_{AB}) - \ln(OR_{CB})$$

$$\mathrm{Var}(\ln OR_{\mathrm{Ind}}) = \mathrm{Var}(\ln OR_{AB}) + \mathrm{Var}(\ln OR_{CB})$$

•Meta analysis with random effect model:

$$\hat{\theta}_i = \theta + \nu_i + \varepsilon_i, \quad \hat{\theta}_i \sim N(\theta, \xi_i^2 + \tau^2)$$

•DerSimonian and Laird (1986): to estimate random variance m_w and standard error of m_w on natural log of OR:

$$m_W = \sum_i w_i y_i / \sum_i w_i, \quad w_i = 1/(t^2 + s_i^2)$$

$$\mathrm{s.e.}(m_W) = 1/\left(\sum_i w_i\right)^{1/2}$$

STATISTICAL ANALYSIS DETAILS (cont.)

$$t^2(\mathrm{DL}) = \max\left\{0, \frac{\left[\sum_i w_{i0}(y_i - y_W(0))^2\right] - (k-1)}{\left[\sum_i w_{i0} - \sum_i w_{i0}^2 / \sum_i w_{i0}\right]}\right\},$$

$$y_W(0) = \sum_i w_{i0} y_i / \sum_i w_{i0}, \text{ and } w_{i0} = 1/s_i^2$$

RESULTS

	Treatments Compared	Odds Ratio	Odds Ratio 95% CI	Nominal P-value
ARISTOTLE	A vs. W	0.89	(0.79, 1.00)	0.0458
Hart et.al 2007	W vs. P	0.74	(0.57, 0.97)	
Indirect Comparison 1	A vs. P	0.66	(0.50, 0.88)	0.004

	Treatments Compared	Odds Ratio	Odds Ratio 95% CI	Nominal P-value
AVERROES	A vs. ASA	0.78	(0.60, 1.01)	0.0555
Hart et.al 2007	ASA vs. P	0.86	(0.69, 1.07)	
Indirect Comparison 2	A vs. P	0.67	(0.48, 0.94)	0.02

	Treatments Compared	Odds Ratio	Odds Ratio 95% CI	Nominal P-value
Meta Analysis Combining Two Indirect Comparisons	A vs. P	0.66	(0.53, 0.82)	0.0002

$t^2(\mathrm{DL})$ = 0 and the p-value for heterogeneity is 0.94 which is not significant

CONCLUSION AND DISCUSSION

•In this mata analysis, apixaban reduced all-cause death comparing to placebo or control
- •Each indirect comparison shows that apixaban reduced all-cause death by at least 33% when compared to placebo or control with a 95% CI with upper bound < 1
- •Apixaban effect is highly consistent in two indirect comparisons
- •This meta analysis combining two indirect comparisons shows that apixaban reduced all-cause death by 34% when compared to placebo/control with a 95% CI with upper bound < 1

•Caveat:
- •The result depends on the assumption of similarity of treatment effect in indirect comparison, which is not testable
- •Only two trials were used to assess between study variation in this meta analysis

•Future Research:
- •Bayesian network with more complex models

REFERENCES

1. Granger C.B, et.al Apixaban versus Warfarin in Patients with Atrial Fibrillation, *The New England Journal of Medicine*, 10.1056/nejmoa1107039
2. Connolly S.J, et al Apixaban in Patients with Atrial Fibrillation, *The New England Journal of Medicine*, 10.1056/NEJMoa1007432
3. Hart R.G, Pearce L.A, Angullar M.I, Meta-analysis: Antithrombotic to Prevent Stroke in Patients Who Have Nonvalvular Atrial Fibrillation, *Annuals of Internal Medicine*, 2007, 146, 857,- 867
4. Glenny AM, Altman DG, Song F et al. Indirect comparisons of competing interventions. *Health Technol Assess* 2005; 9: 1–134, iii–iv.
5. Song F, Harvey I, Lilford R. Adjusted indirect comparison may be less biased than direct comparison for evaluating new pharmaceutical interventions. *J Clin Epidemiol* 2008; 61: 455–463.
6. Bucher HC, Guyatt GH, Griffith LE, Walter SD. The results of direct and indirect treatment comparisons in meta-analysis of randomized controlled trials. *J ClinEpidemiol 1997; 50: 683–691.*